Investigational Therapy of Alport Syndrome With ELX-02

A. Hariri[1], K. Omachi[2], V. Badarinarayana[1], M. Cox[1], J. Miner[2], V. Modur[1]

[1]Eloxx Pharmaceuticals, Watertown, Massachusetts, USA, [2]Washington University in St Louis, St Louis, Missouri, USA

Background

Alport syndrome is a genetic disease caused by mutations in COL4A3, COL4A4 and COL4A5 genes leading to glomerular fragility, hematuria and proteinuria resulting in ESRD early in life. The most frequent form is X-linked Alport syndrome, which is caused by mutations in COL4A5. ELX-02 is an aminoglycoside analog specifically designed to readthrough premature termination (nonsense) codon mutations and produce full-length proteins instead of truncated proteins. We show here the potential for ELX-02 as an investigational therapy in Alport Syndrome..

Methods

Nonsense mutations in COL4A5 associated with Alport syndrome was compiled from ClinVar, LOVD, Ensemble database. The number of unique nonsense mutations and frequency of each were determined. Type of nonsense codon (UGA, UAG or UAA) was derived from sequence variants. 13 of the mutants were selected for analysis by dual reporter assay for responsiveness to ELX-02 induced readthrough. To assess expected clinical exposure in kidney for ELX-02, a Physiologically Based Pharmacokinetic (PBPK) model was built based on preclinical pharmacokinetic data from mouse, rat and dog studies using Gastroplus simulation software.

Results

Analysis of nonsense mutations compiled across different databases showed approximately 6% of reported pathogenic mutations in COL4A5 are of the nonsense subtype. All the tested mutations had >3-fold increase in readthrough with an average of 13.7-Fold

Compiled COL4A5 variant data from LOVD

Category	Count
Number of individuals	2328
Number of variants	2325
Number of substitution mutations	1771
Number of nonsense mutations	98

Types of variants detected in COL4A5

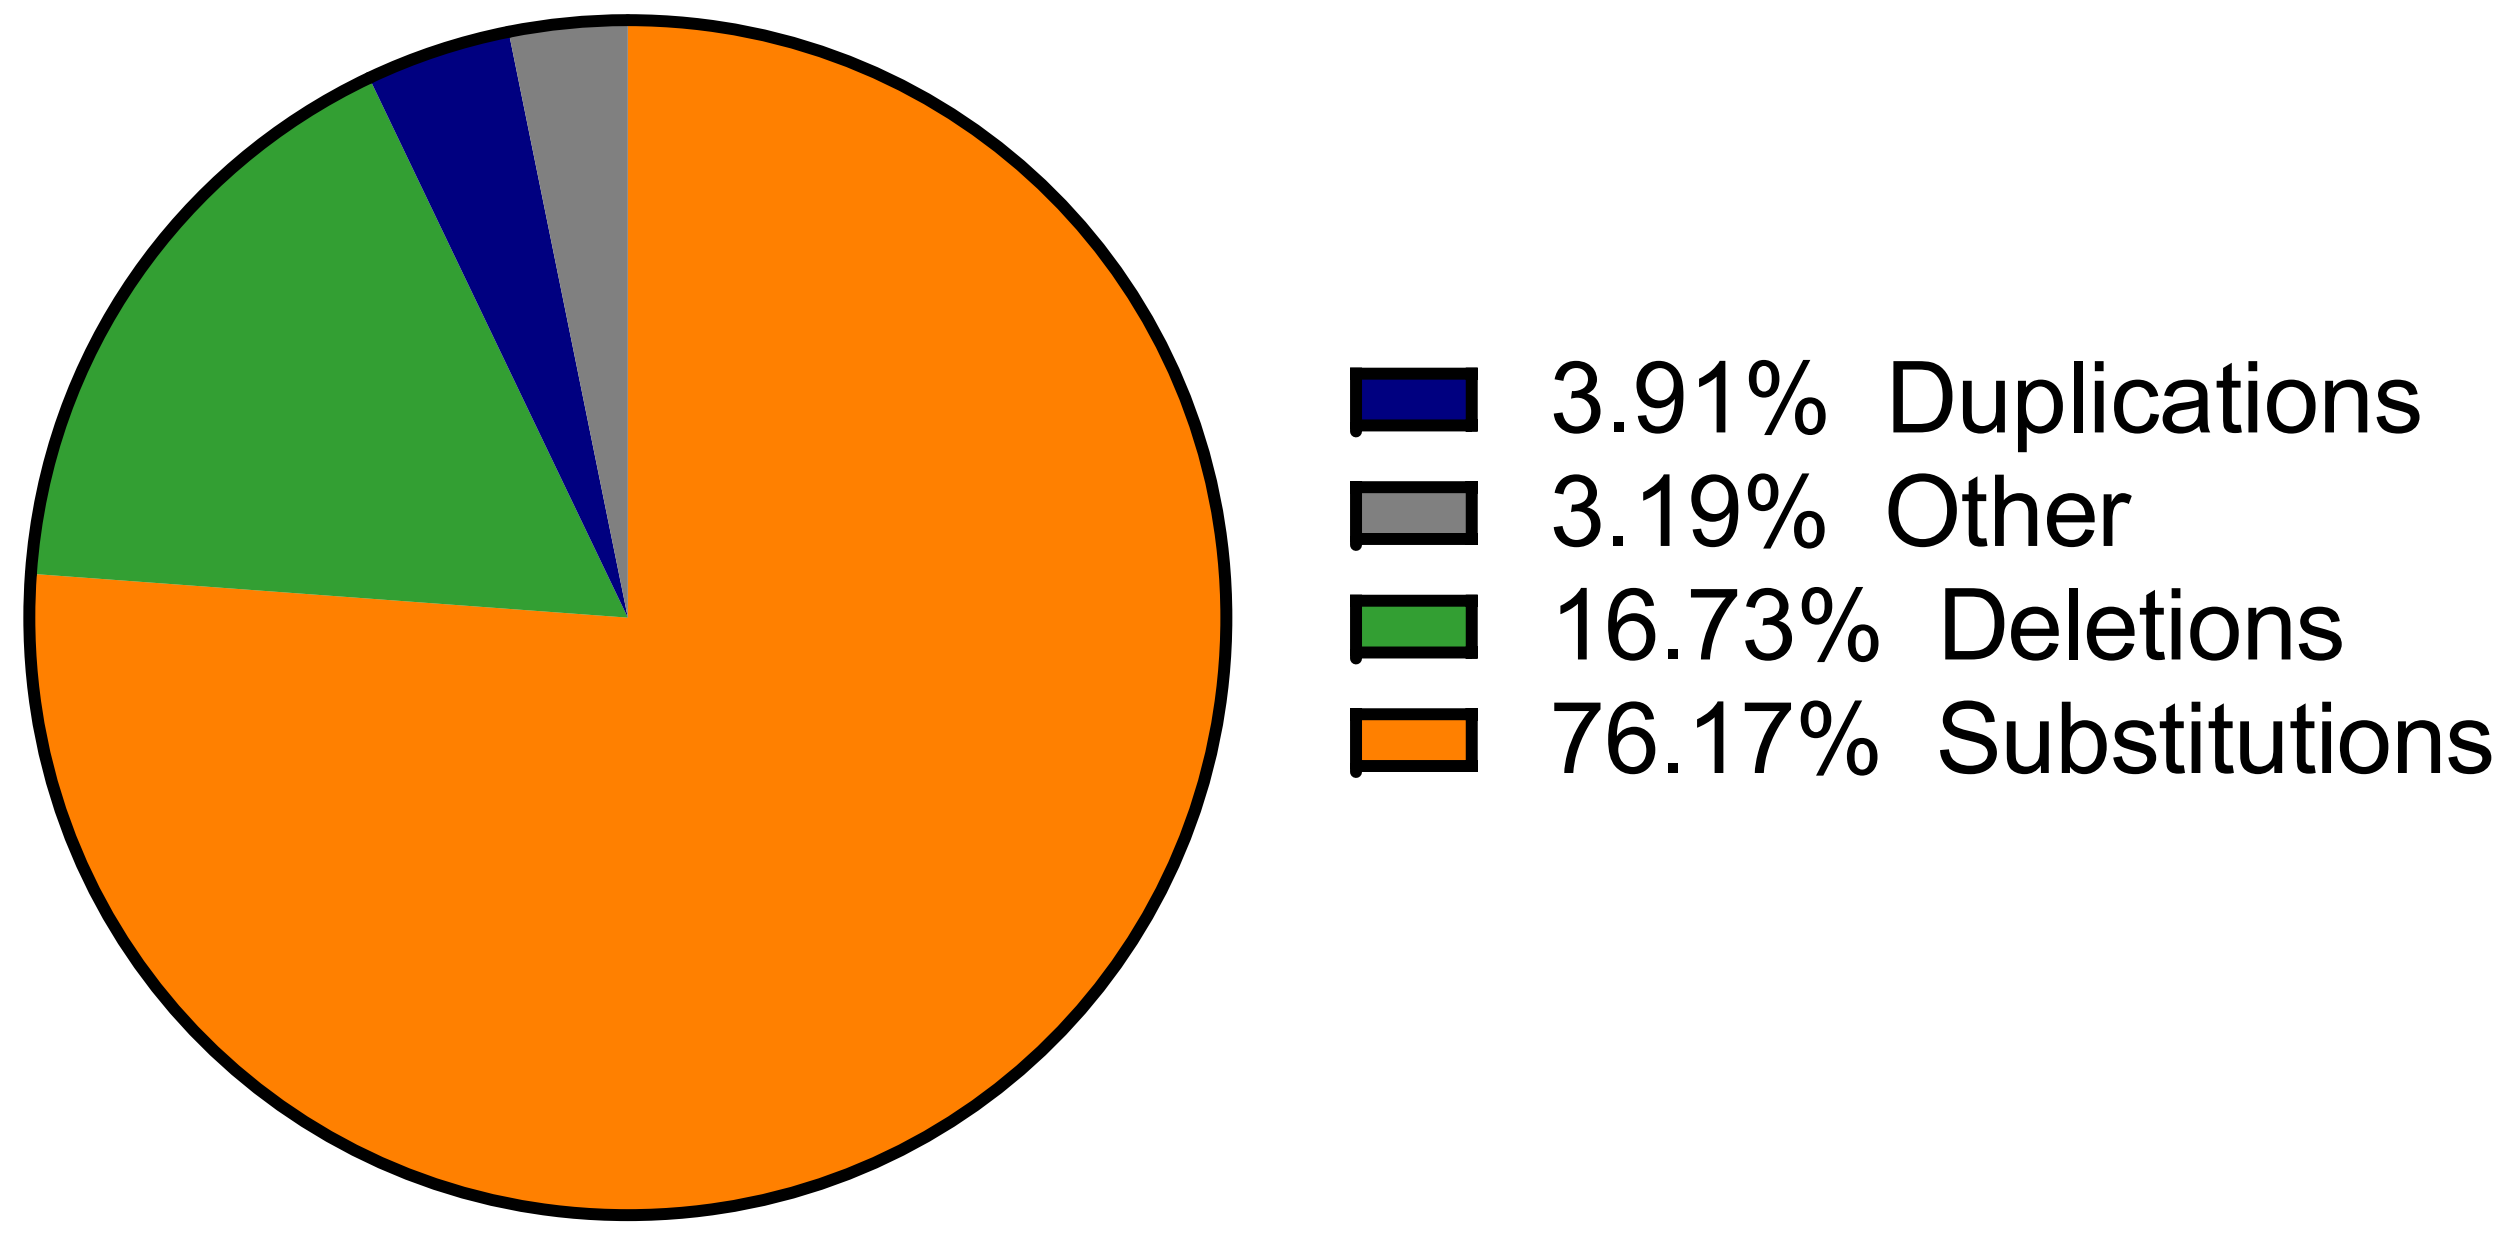
3.91% Duplications
3.19% Other
16.73% Deletions
76.17% Substitutions

Frequency of Stop codons detected in COL4A5

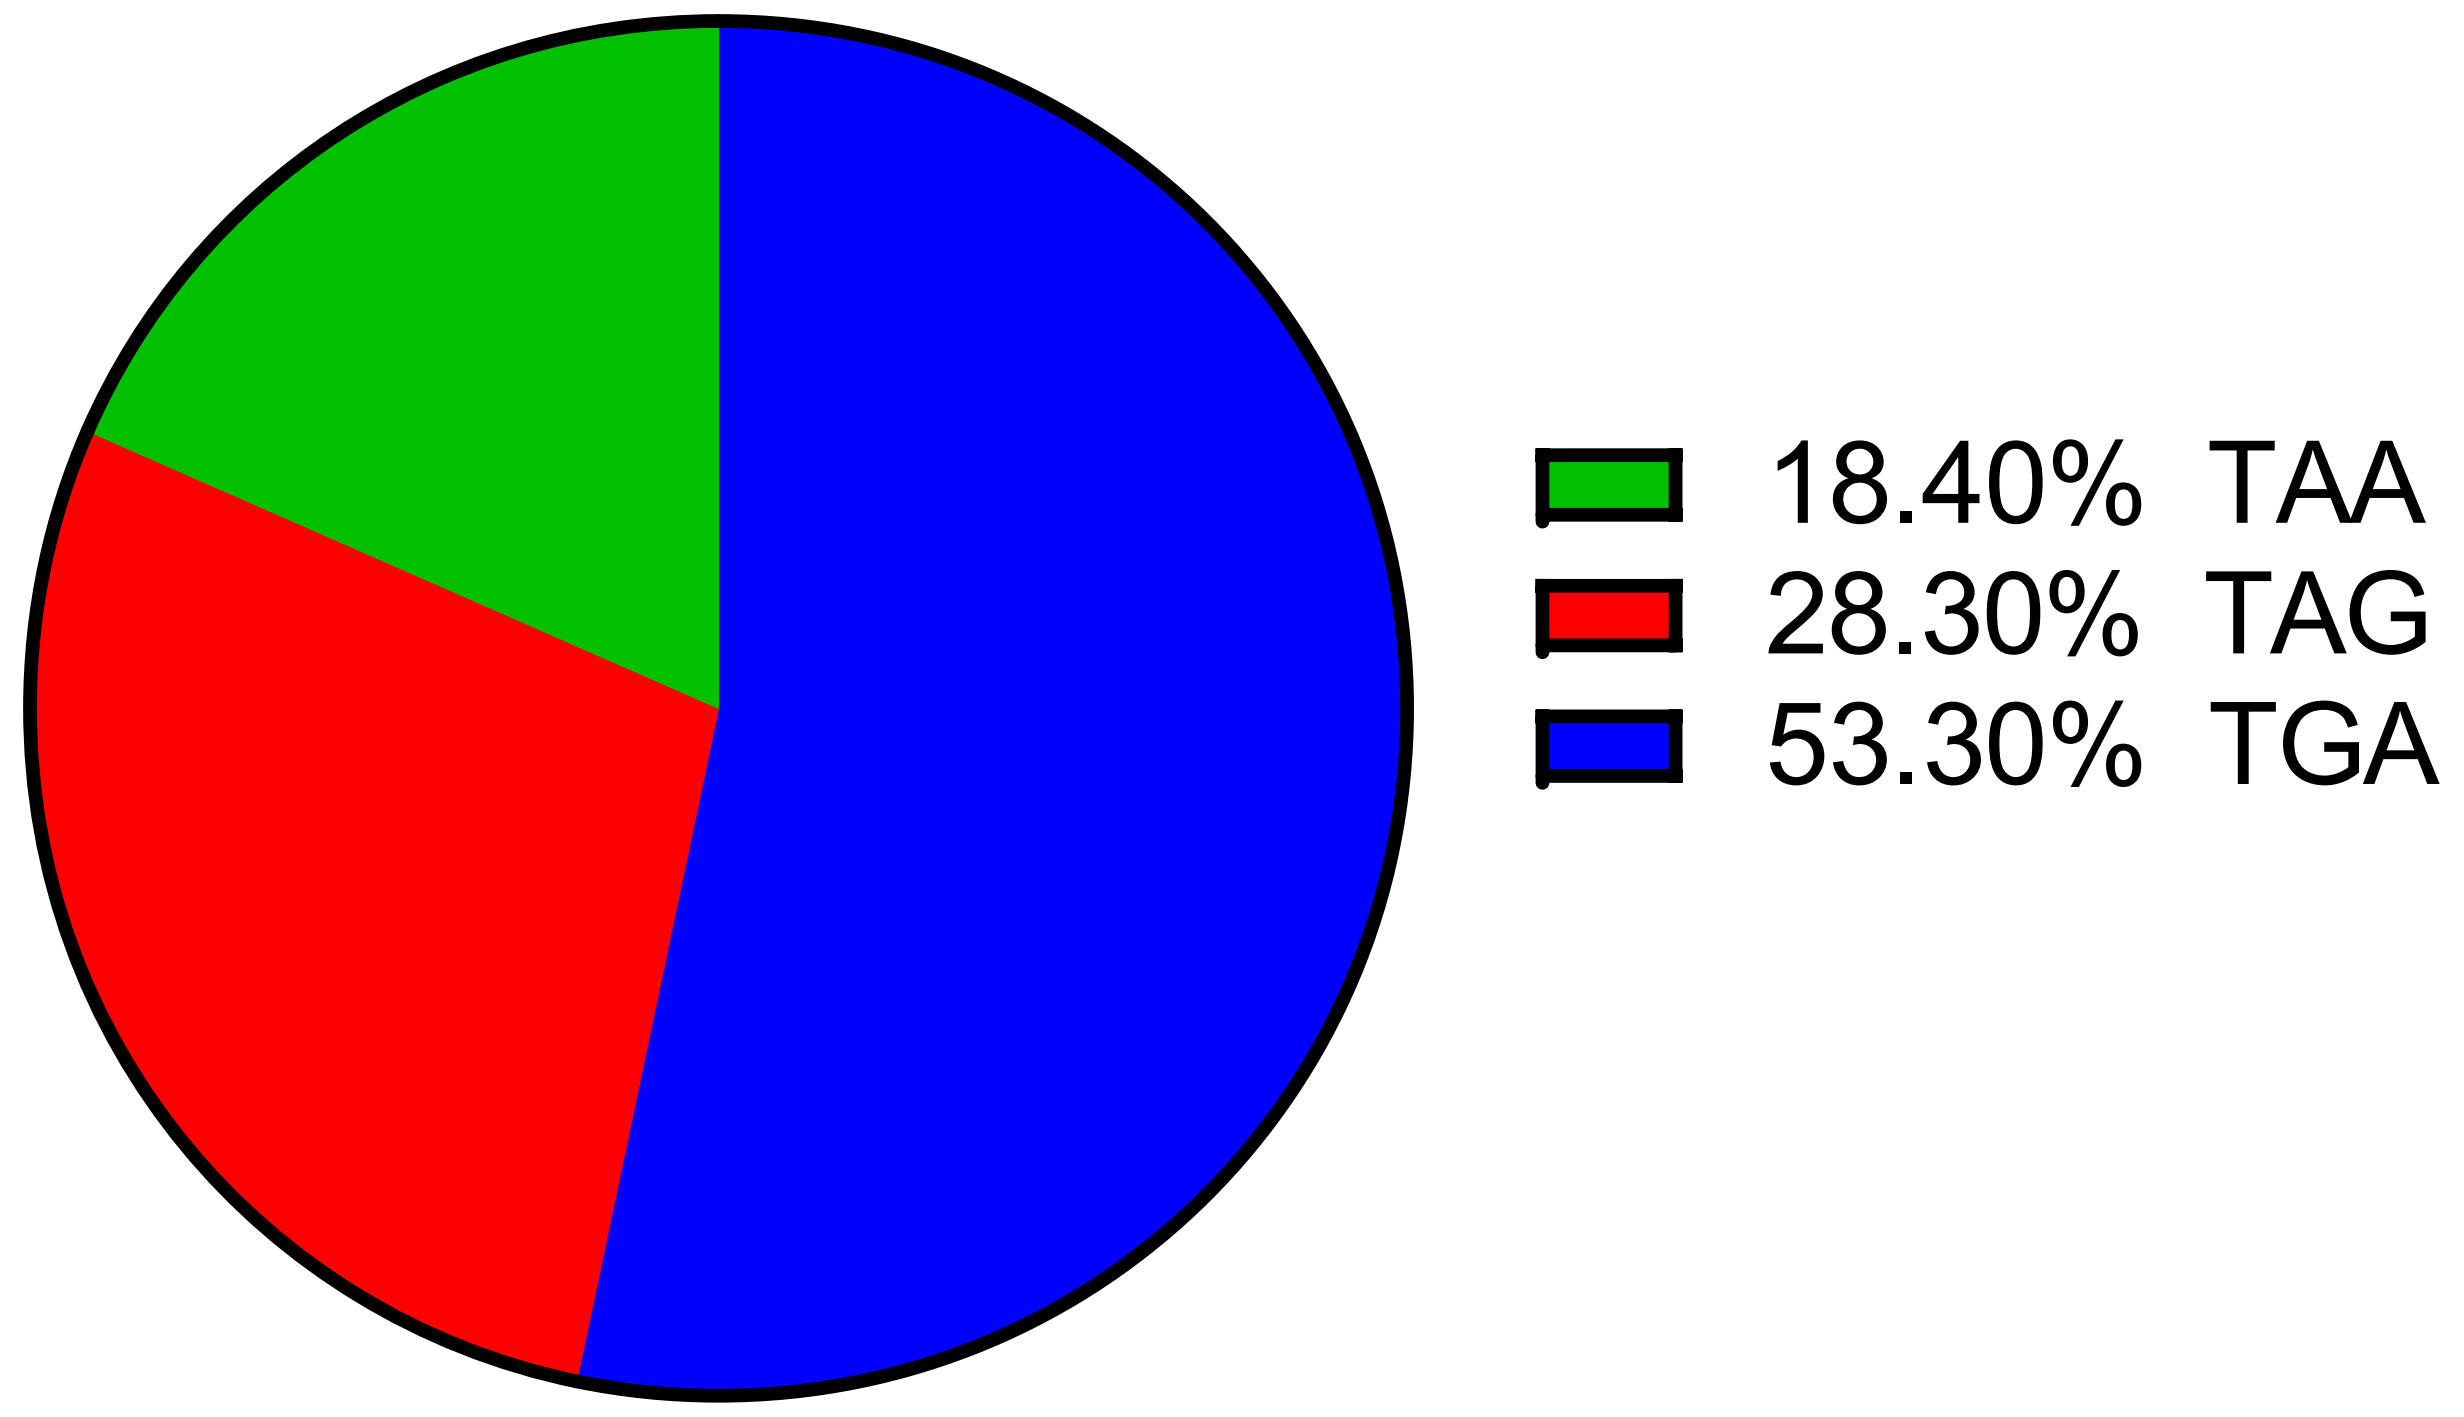
18.40% TAA
28.30% TAG
53.30% TGA

Analysis of read-through activity across COL4A5 PTCs using Dual reporter assays

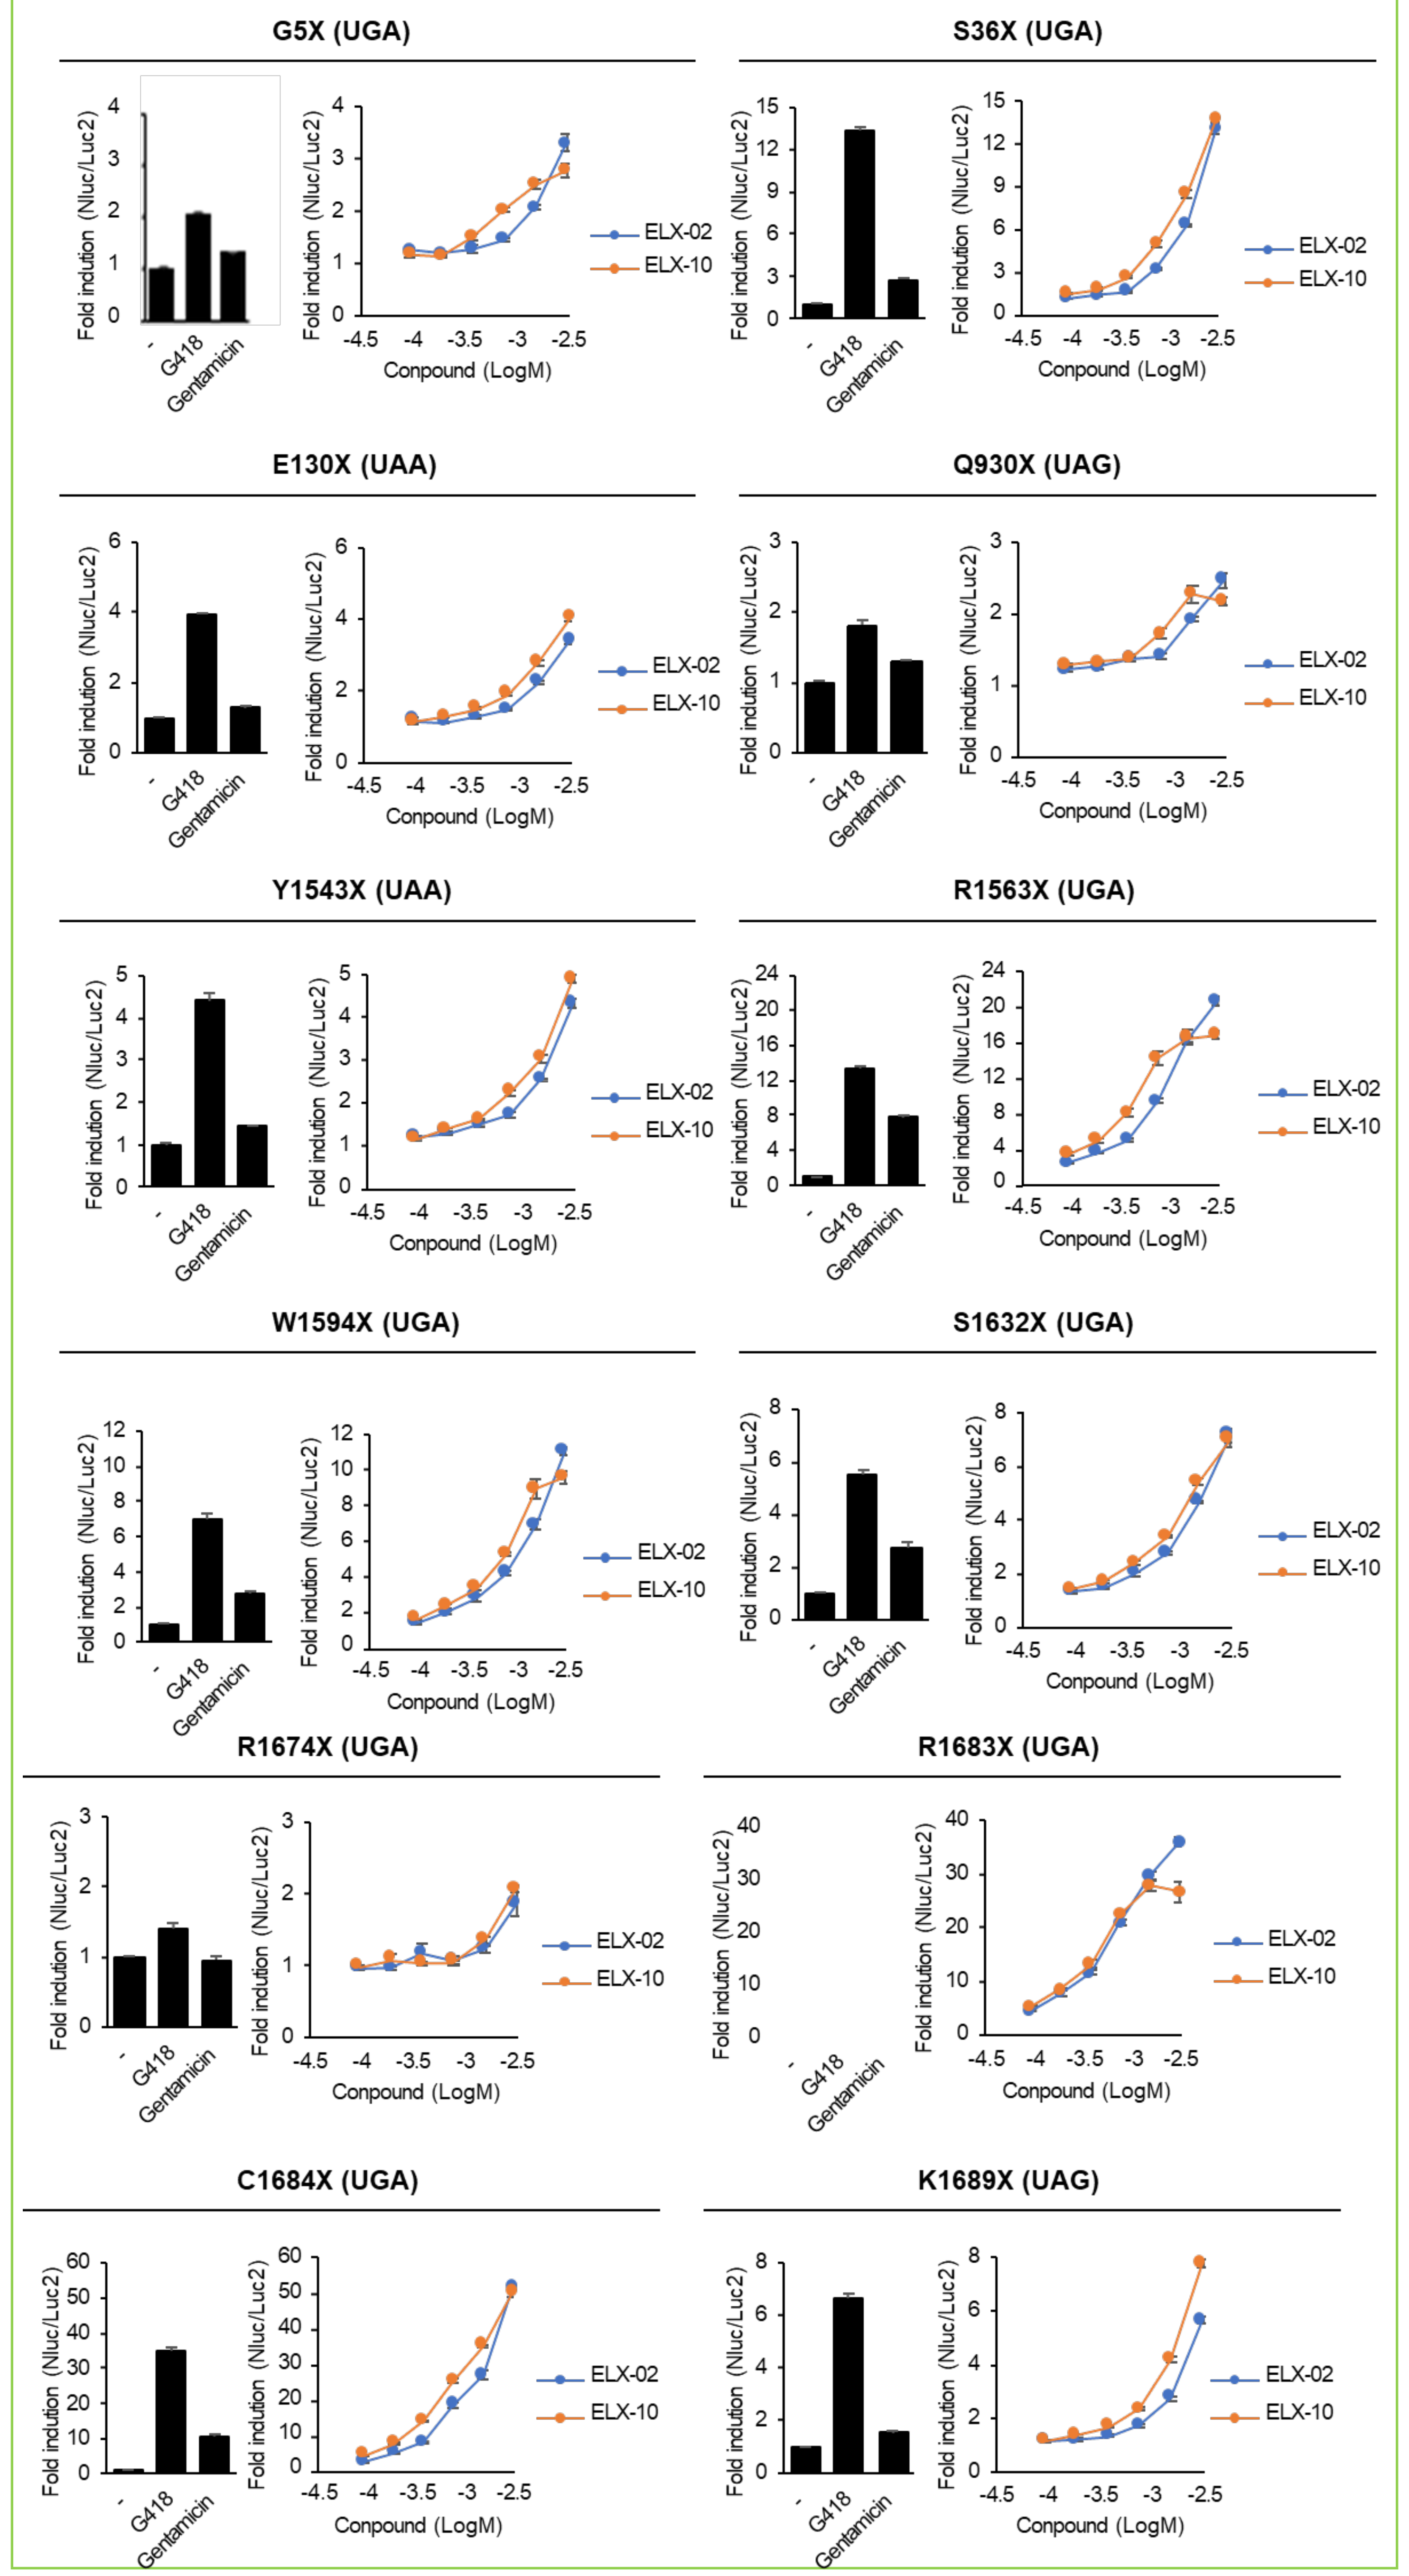
G5X (UGA)
S36X (UGA)
E130X (UAA)
Q930X (UAG)
Y1543X (UAA)
R1563X (UGA)
W1594X (UGA)
S1632X (UGA)
R1674X (UGA)
R1683X (UGA)
C1684X (UGA)
K1689X (UAG)
Fold indution (Nluc/Luc2)
G418
Gentamicin
Conpound (LogM)
ELX-02
ELX-10

PBPK modeling

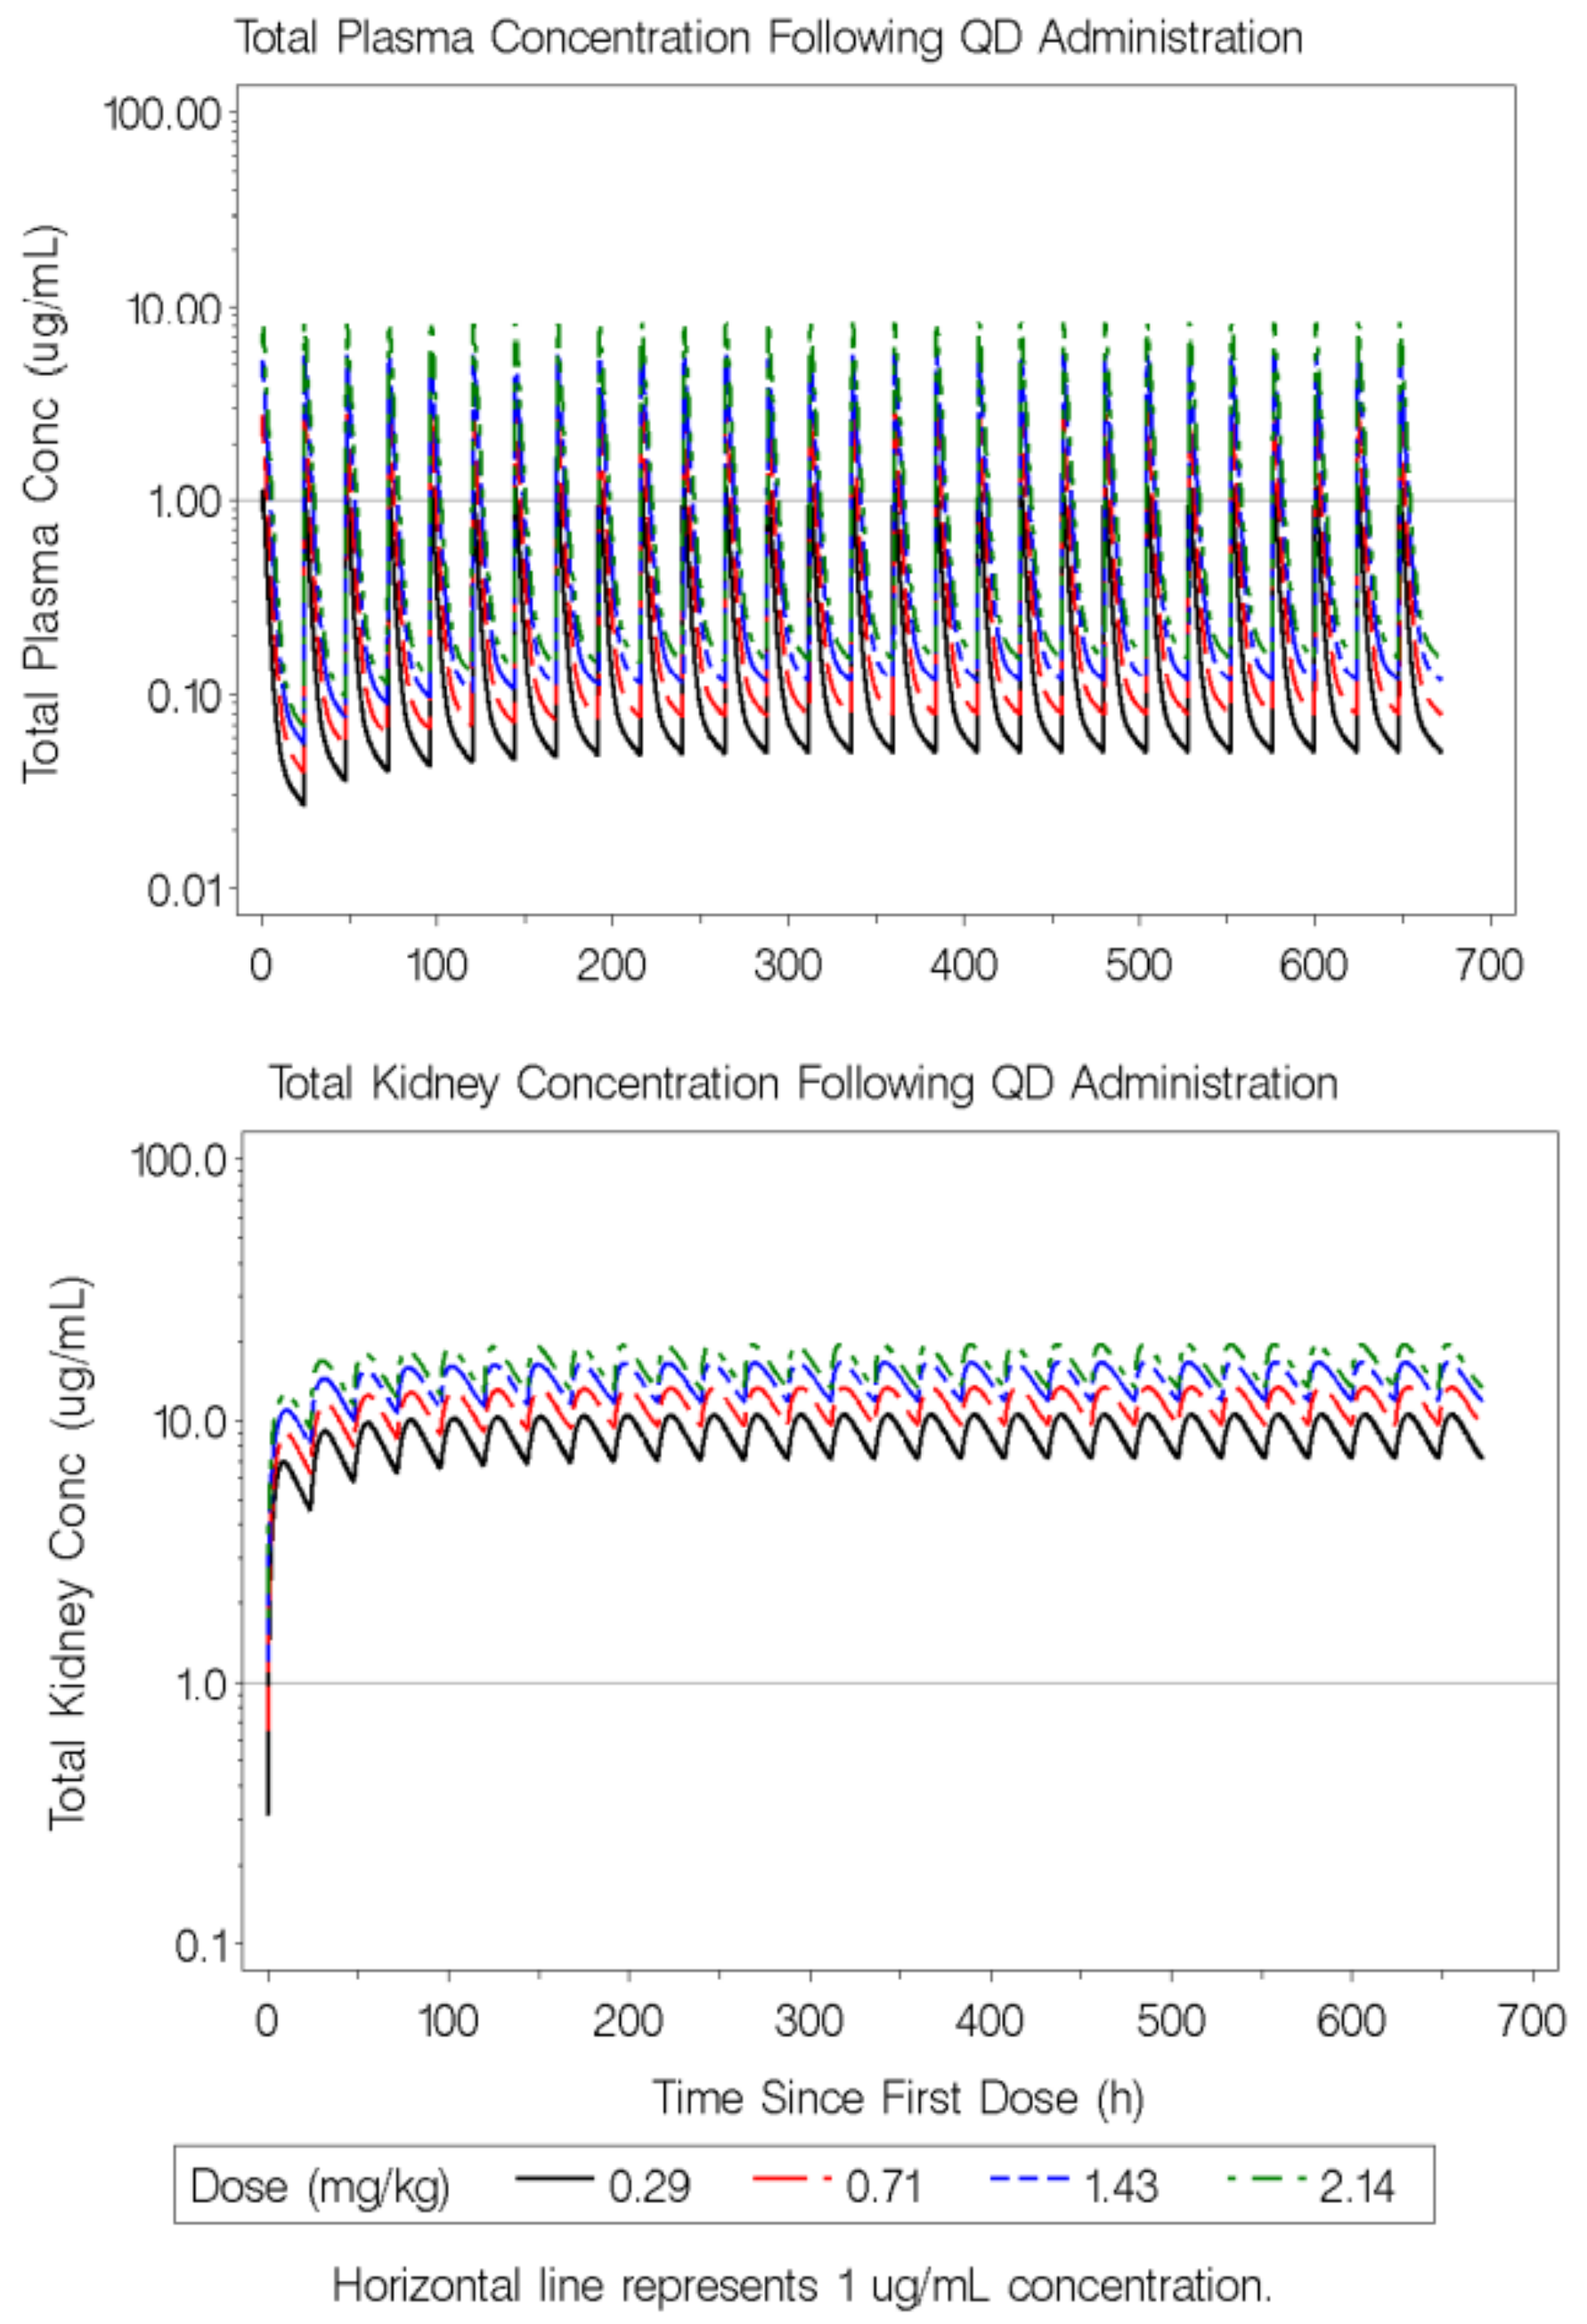
Total Plasma Concentration Following QD Administration
Total Plasma Conc (ug/mL)
Total Kidney Concentration Following QD Administration
Total Kidney Conc (ug/mL)
Time Since First Dose (h)
Dose (mg/kg)
0.29
0.71
1.43
2.14
Horizontal line represents 1 ug/mL concentration.

PBPK modeling showed that ELX-02 levels in the kidney are >50-fold greater than plasma levels across a range of doses suggesting high exposures in the kidneys even at 0.71 mg/kg dose

Conclusion

The promise of this investigational therapy in Alport syndrome is supported by several lines of evidence. 1) Nonsense mutations in COL4A5 are a significant cause of Alport Syndrome. 2) ELX-02 shows nonsense mutation readthrough across a range of nonsense mutations. 3) PBPK modeling shows high levels of ELX-02 exposures can be achieved in the kidneys even at doses below those currently being used in clinical trials.

Acknowledgements: Anupama Reddy for sequence analysis