Identification, validation and initial SAR exploration of oncoribosome targeting macrolides

*Wesley F. Austin**, *Simran Padhye, Shruti Apte, Vasu Badarinarayana, Roger B. Clark, Saleh Rashed, Shuhao Shi, Esteban Terzo, and Christy Wang.*

Eloxx Pharmaceuticals, 480 Arsenal Way, Watertown, MA 02472

Background:

The transformation of normal cells into cancerous cells is associated with changes in the structure and function of ribosomes. This includes an increase in biogenesis, development of mutations, and alterations in post-translational modifications of ribosomal proteins.[1] These "oncoribosomes" show shifts in the pattern of mRNA translation compared to normal ribosomes, which can lead to the genesis or sustainment of tumors.[2] Therefore, we believe that selectively targeting oncogenic ribosome forms is a potential anti-tumor therapy. To achieve this, we are leveraging our knowledge on the synthesis of ribosomal modulating agents (RMAs) and their unique interactions.[3] We have prepared a library of >1000 RMAs and screened this library against several cancerous cell lines as well as non-cancerous controls.

Assay methods:

Cell Culture: HEK293 (human embryonic kidney) cells used as counter-screen. SW1417 and SW948 are colorectal cancer derived cell lines. NICH446 are small-cell lung cancer cells. NCIH1693 are non-small-cell lung cancer cells. DAUDI are Burkitts lymphoma cancer cells. All cell lines grown as recommended by ATCC.

Screen: Cell lines were treated with compounds for 72 (HEK293), 86 (NCIH446), 120 (NCIH1693, DAUDI) and 144 (SW1417) hours. Cell viability was quantitated by assessing ATP levels at the end of the assay.

Organoids: CRC patient-derived organoids and a normal-tissue derived one were treated with 9 doses of compound 9 at 60uM top concentration for 7 days. Cell viability was quantitated by assessing ATP levels at the end of the assay.

Xenograft: Mice were inoculated subcutaneously with SW948 cells for tumor development and treated with 50mpk QD of compound 9, administered orally, for 22 days.

Protein translation detection: SW1417 cells were seeded for 48 hours and subsequently treated with compounds 9 and 11 for 48 hours for both assays.

1. Nascent protein translation was assessed using a metabolic labeling assay that incorporates a methionine analog, which was fluorescently labeled on a 96-well plate and quantitated.[4]

2. p21 protein expression was assessed via Western Blot assay. Protein lysates from treated cells were loaded on 4-12% acrylamide gels. Anti-p21 antibody was used to detect protein on gel.[4] Image studio 5.2 software was used to acquire images and quantitate intensities from both protein translation detection assays. GraphPad Prism 9 software was used to calculate tumor volume, IC50 (uM), and relative protein expression. Spotfire (TIBCO) 12.0.0.223 was used to plot bar graph for organoid response.

Compounds selective for cancerous cell lines are present

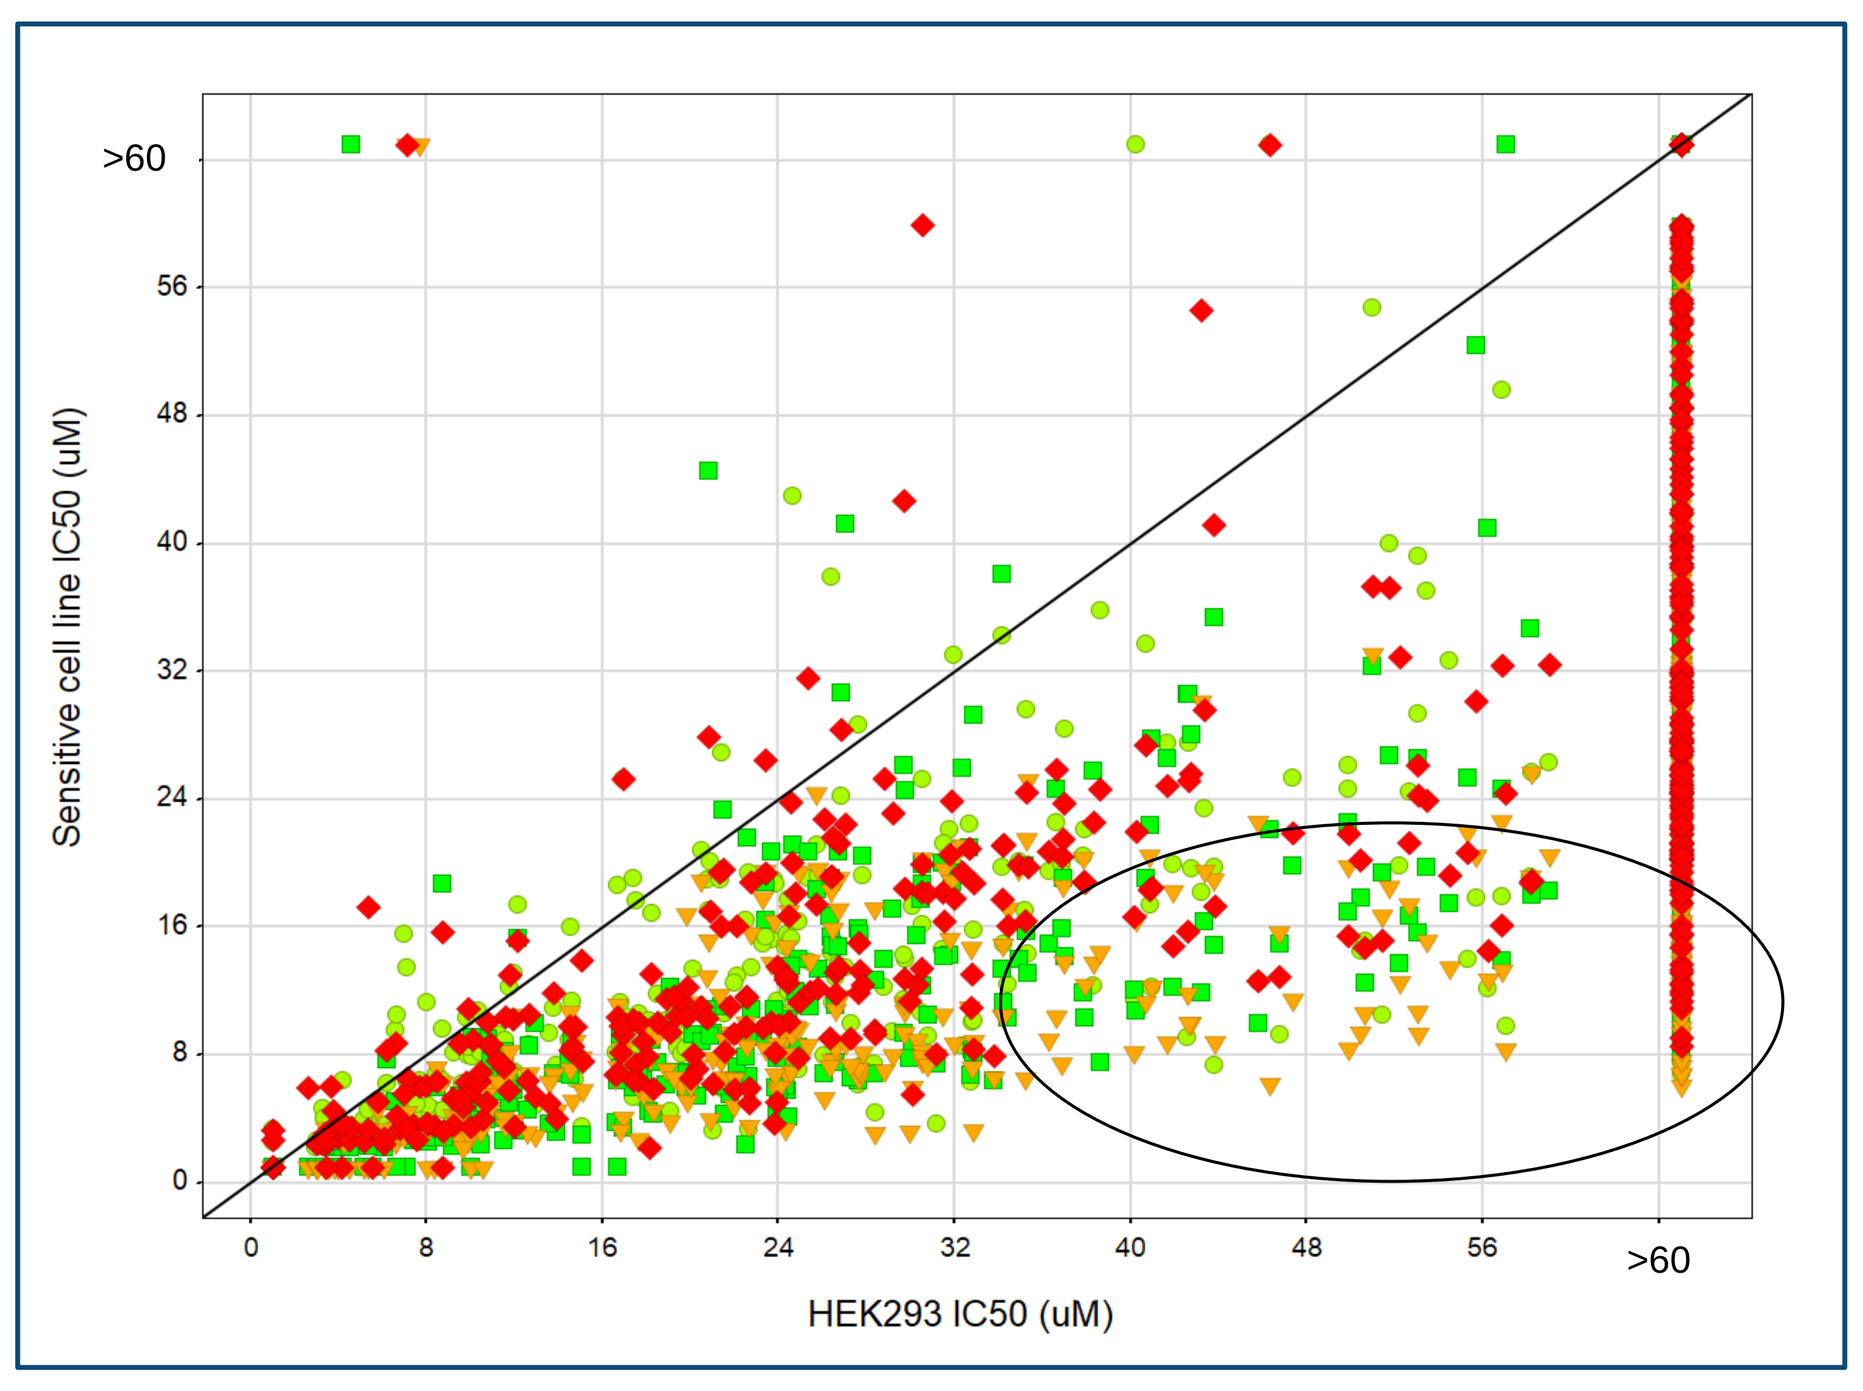
>60
56
48
40
32
24
16
8
0
Sensitive cell line IC50 (uM)
0
8
16
24
32
40
48
56
>60
HEK293 IC50 (uM)
DAUDI
NCI-H446
SW-1417
NCI-H1693

Aryl and lipophilic compounds are more potent

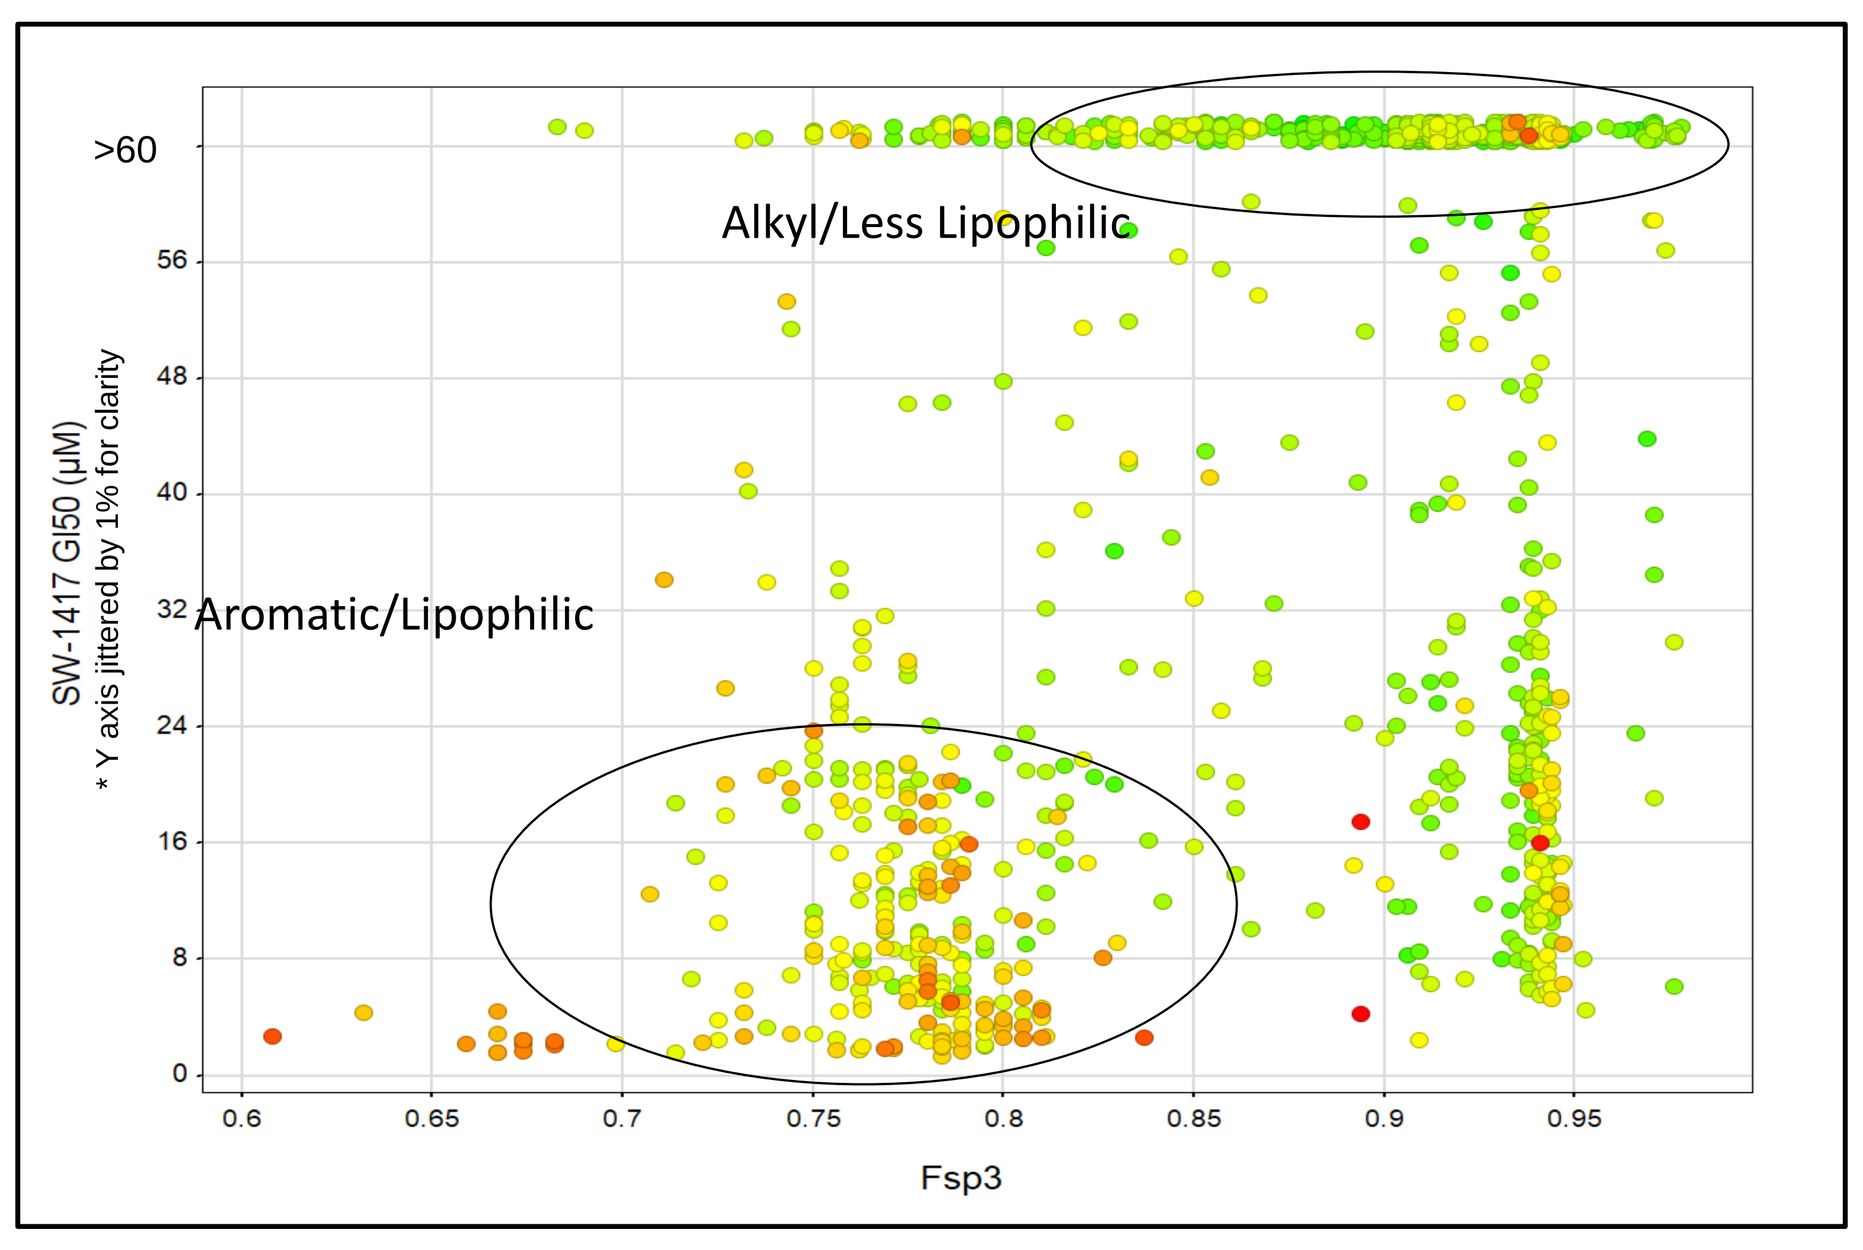
>60
56
48
40
32
24
16
8
0
Alkyl/Less Lipophilic
Aromatic/Lipophilic
SW-1417 GI50 (µM)
* Y axis jittered by 1% for clarity
0.6
0.65
0.7
0.75
0.8
0.85
0.9
0.95
Fsp3
logP
0.77
4.54
8.32

SAR Examination of an example series

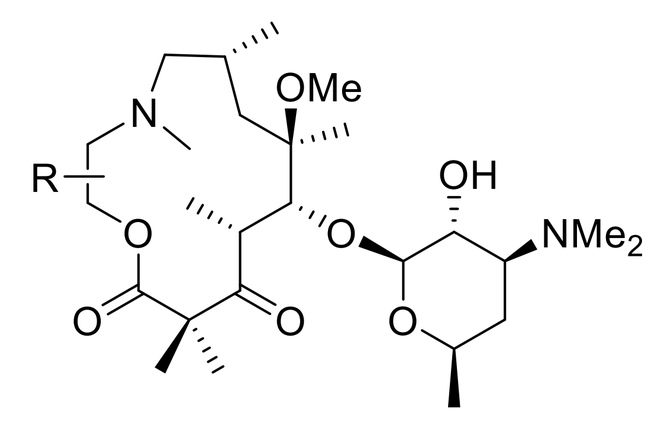
R
N
OMe
OH
NMe2
O
O
O
O
O

R	#	GI50 (µM) DAUDI	NCI-H446	SW-1417	NCI-H1693	HEK293	AUC of inhibition curve (normalized) DAUDI	NCI-H446	SW-1417	NCI-H1693	HEK293	% at Max Response (%) DAUDI	NCI-H446	SW-1417	NCI-H1693	HEK293
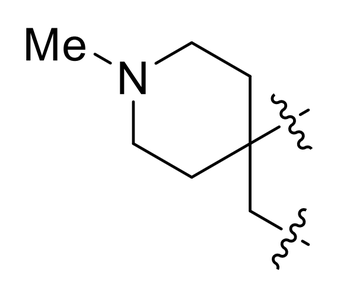	**1**	>60	>60	>60	>60	>60	0.07	0.13	0.07	0.07	0.01	86	68	64	73	95
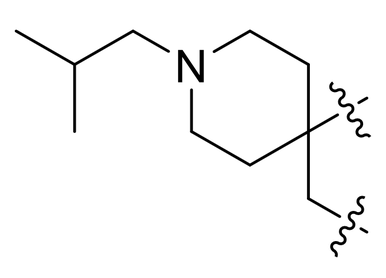	**2**	39.9	25.9	28.7	>60	>60	0.38	0.45	0.27	0.17	0.07	7	-25	27	61	75
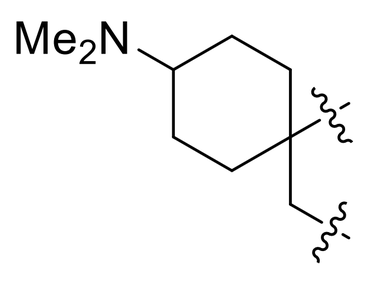	**3**	>60	>60	>60	>60	>60	0.00	0.06	0.01	0.00	0.00	100	89	78	99	99
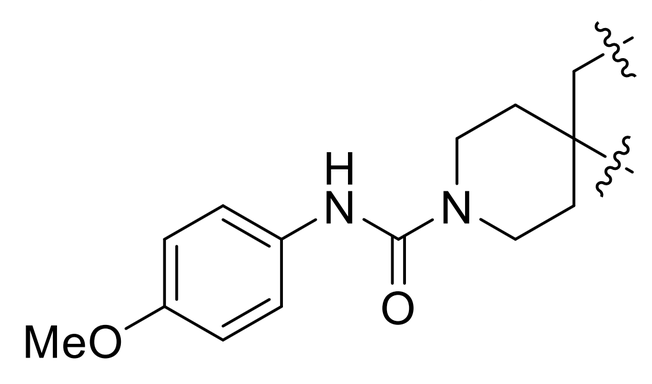	**4**	>60	20.2	20.5	>60	>60	0.09	0.42	0.30	0.07	0.00	76	18	-10	58	98
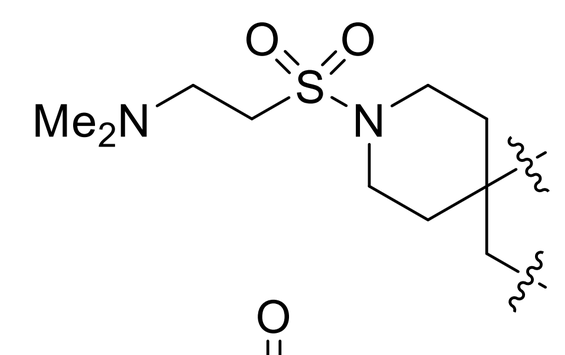	**5**	20.4	13.6	12.2	>60	>60	0.56	0.58	0.40	0.17	0.05	7	-27	-5	59	85
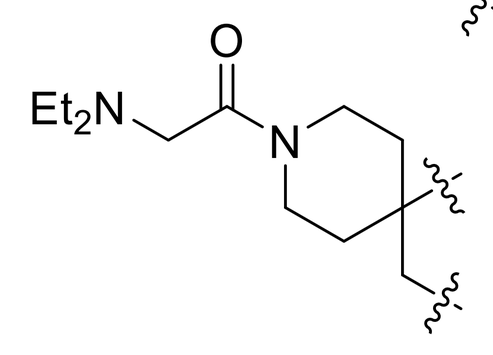	**6**	>60	>60	>60	>60	>60	0.00	0.00	0.00	0.00	0.00	104	93	87	104	90
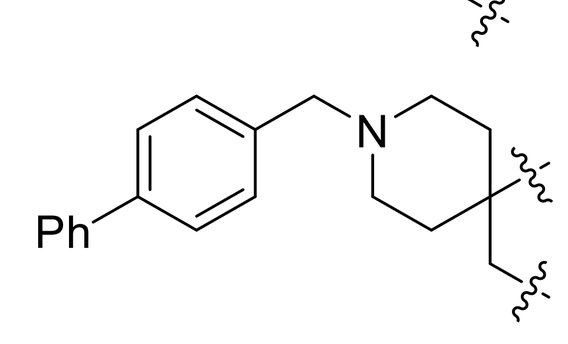	**7**	<2	<2	<2	<2	<2	0.96	0.96	0.96	0.96	0.92	-64	-106	-102	-109	-83

Benzyl and homobenzyl piperidines are most interesting

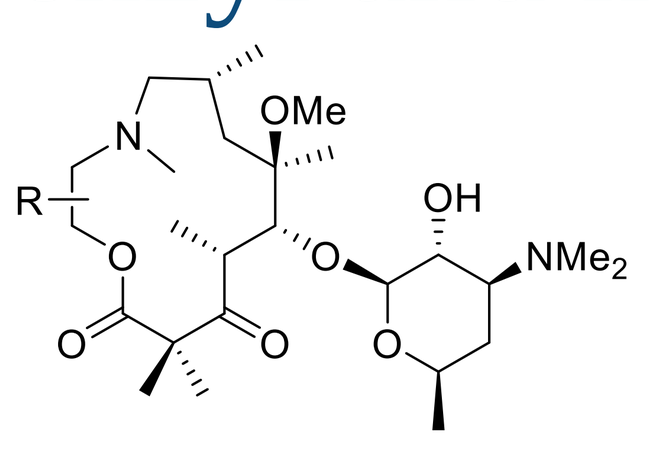
R
N
OMe
OH
NMe2
O
O
O
O
O

R	#	GI50 (µM) DAUDI	NCI-H446	SW-1417	NCI-H1693	HEK293	AUC of inhibition curve (normalized) DAUDI	NCI-H446	SW-1417	NCI-H1693	HEK293	% at Max Response (%) DAUDI	NCI-H446	SW-1417	NCI-H1693	HEK293
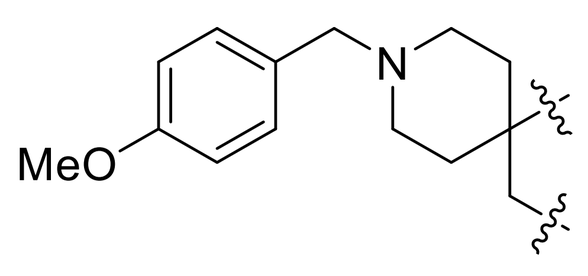	**8**	29.1	23.4	10.1	15.0	>60	0.40	0.45	0.48	0.58	0.10	25	-4	0	-46	64
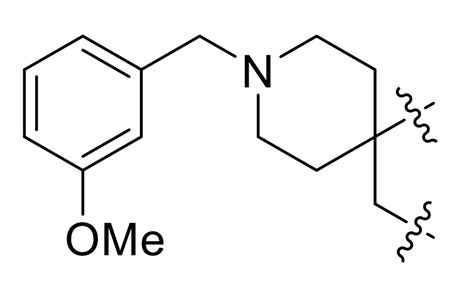	**9**	19.9	22.1	14.1	11.4	58.2	0.48	0.53	0.48	0.67	0.20	24	-16	-24	-39	47
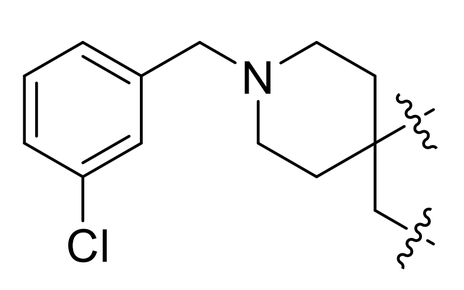	**10**	9.6	6.7	2.2	4.4	15.7	0.71	0.79	0.79	0.83	0.59	3	-57	-80	-61	1
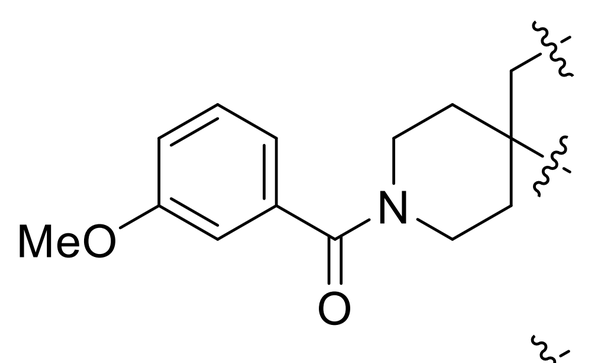	**11**	>60	>60	>60	>60	>60	0.03	0.16	0.09	0.12	N/D	87	70	67	66	95
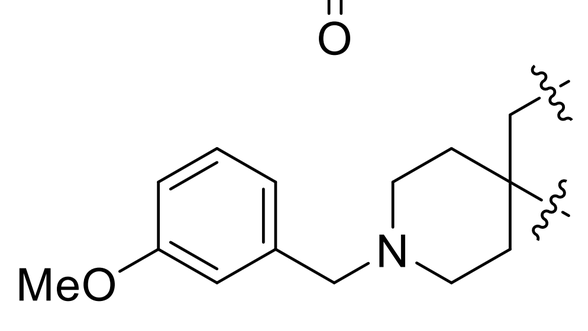	**12**	13.1	10.4	4.8	7.9	19.3	0.70	0.79	0.78	0.80	0.64	1	-79	-89	-83	-80
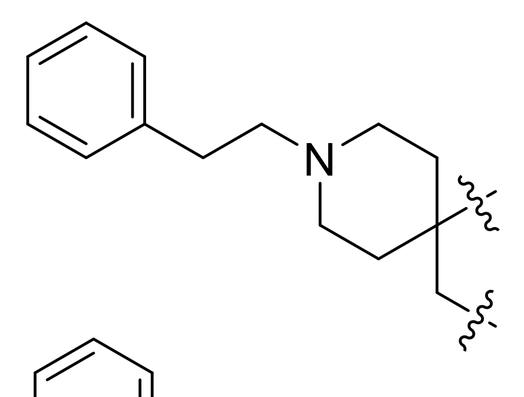	**13**	12.3	18.4	8.0	8.3	36.1	0.69	0.53	0.64	0.73	0.29	6	-22	-53	-57	13
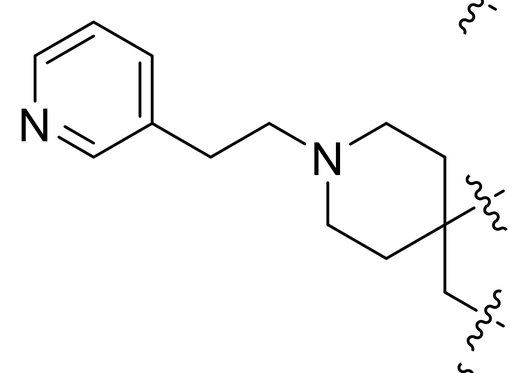	**14**	>60	>60	27.7	>60	>60	0.04	0.16	0.28	0.15	0.02	86	74	35	67	87
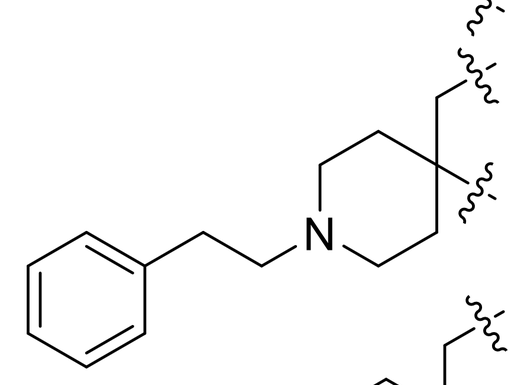	**15**	6.3	8.2	>60	6.7	13.4	0.83	0.80	0.73	0.81	0.65	2	-59	-78	-63	-7
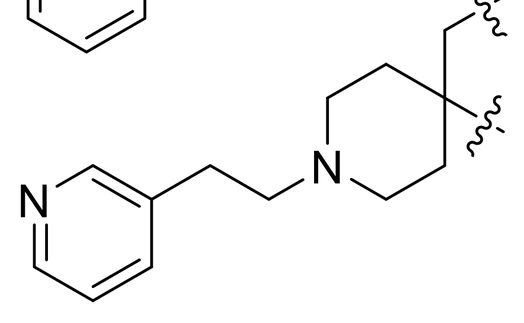	**16**	53.6	>60	13.1	47.9	>60	0.18	0.19	0.42	0.27	0.08	39	48	20	32	78

Compound 9 inhibits protein translation in CRC cell lines

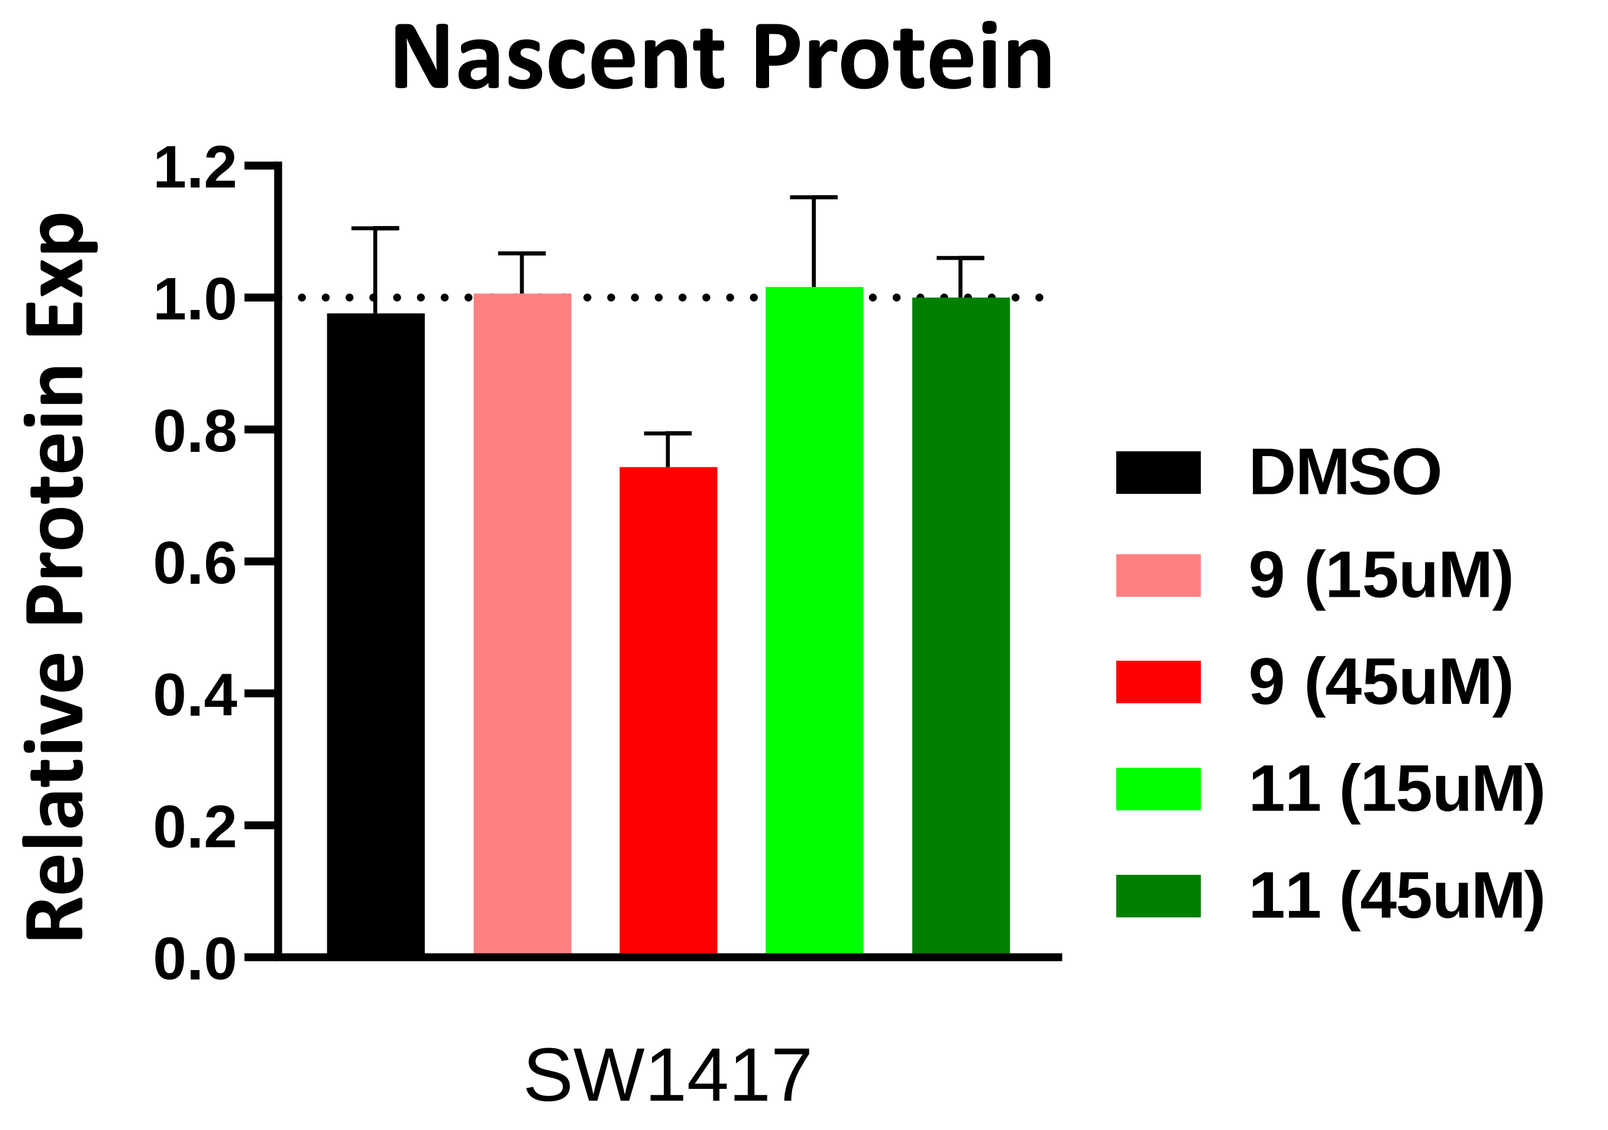
Nascent Protein
Relative Protein Exp
1.2
1.0
0.8
0.6
0.4
0.2
0.0
SW1417
DMSO
9 (15uM)
9 (45uM)
11 (15uM)
11 (45uM)

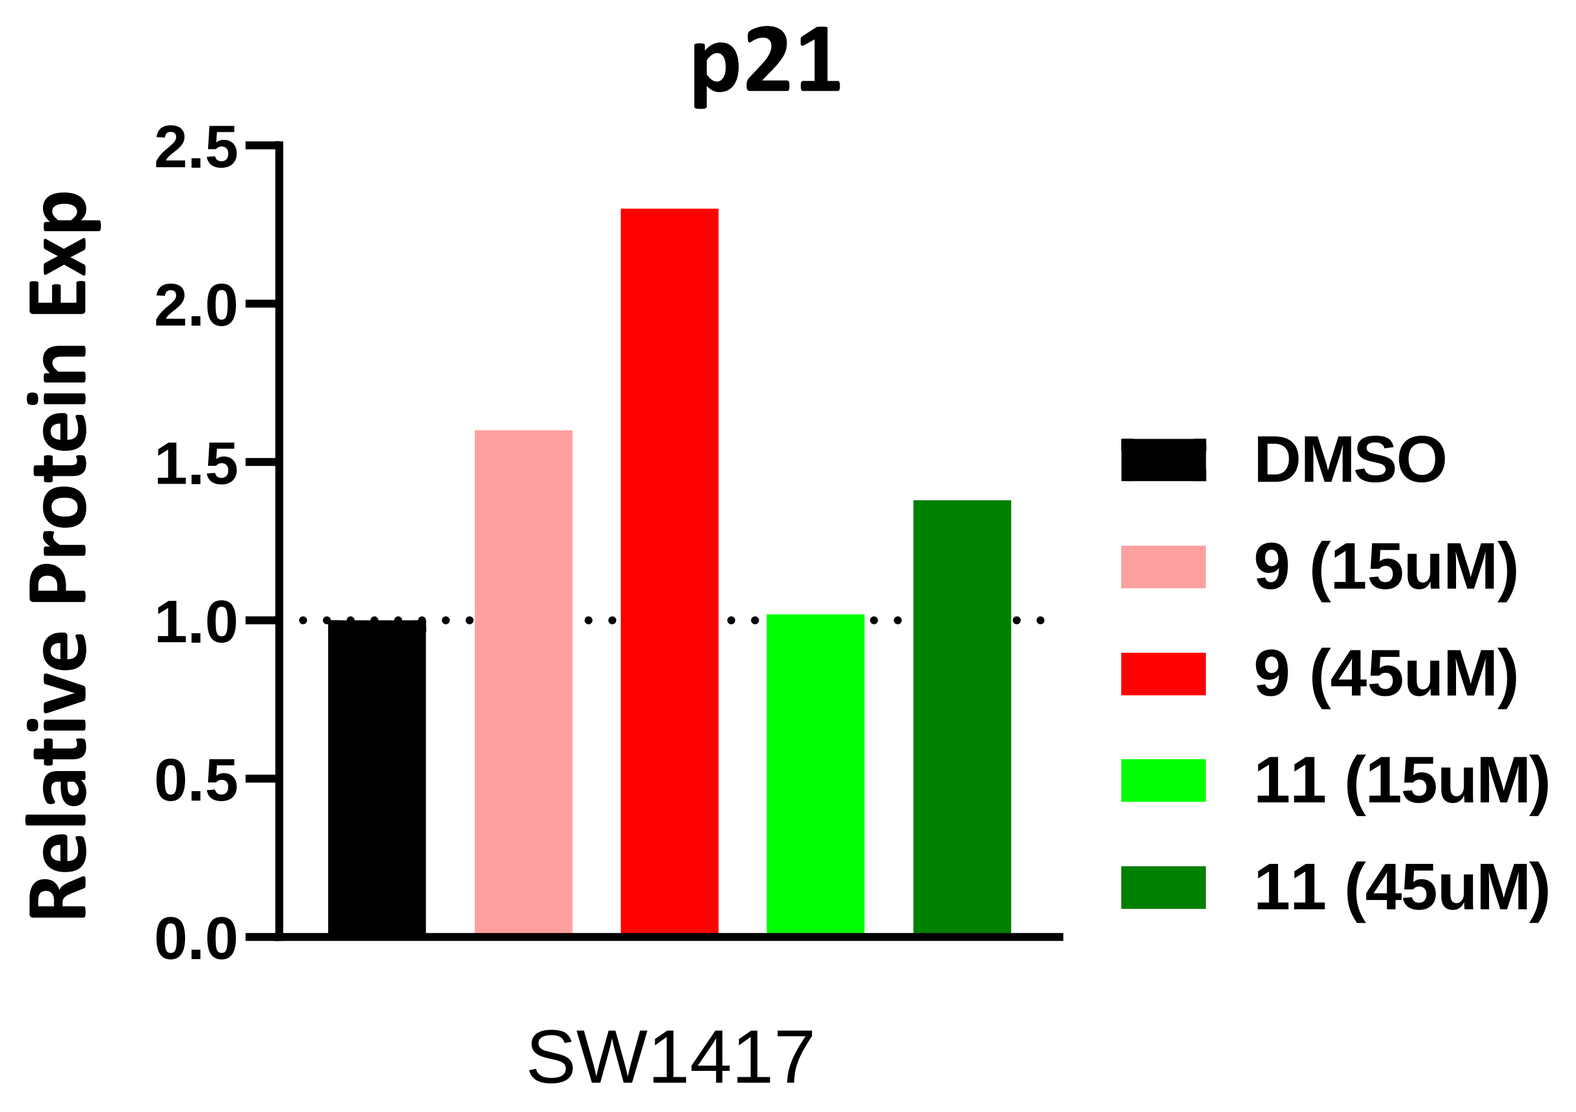
p21
Relative Protein Exp
2.5
2.0
1.5
1.0
0.5
0.0
SW1417
DMSO
9 (15uM)
9 (45uM)
11 (15uM)
11 (45uM)

Left panel: Compound 9 targets oncoribosomes of a CRC sensitive cell line (SW1417), which inhibits translation of selective subsets of protein resulting in their decreased abundance. Compound 11, which shows no growth inhibition, does not.

Right panel: Cell cycle regulator p21 is known to be upregulated after ribosome biogenesis stress, leading to cell cycle arrest and subsequent apoptosis.

Compound 9 selectively inhibits CRC organoids and causes tumor growth inhibition

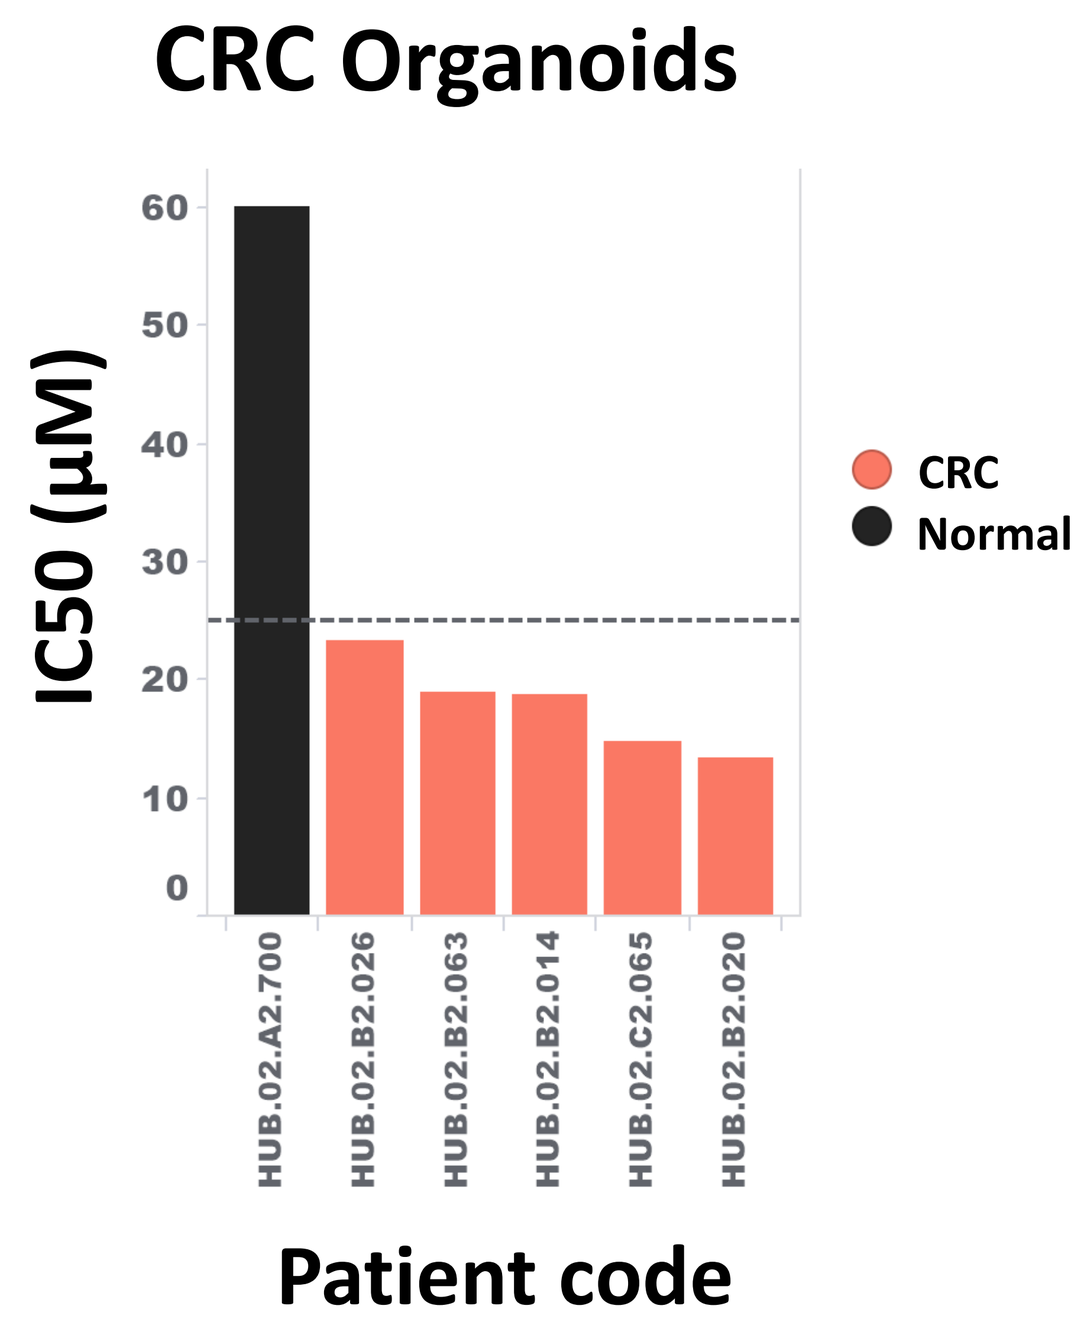
CRC Organoids
IC50 (µM)
60
50
40
30
20
10
0
HUB.02.A2.700
HUB.02.B2.026
HUB.02.B2.063
HUB.02.B2.014
HUB.02.C2.065
HUB.02.B2.020
CRC
Normal
Patient code

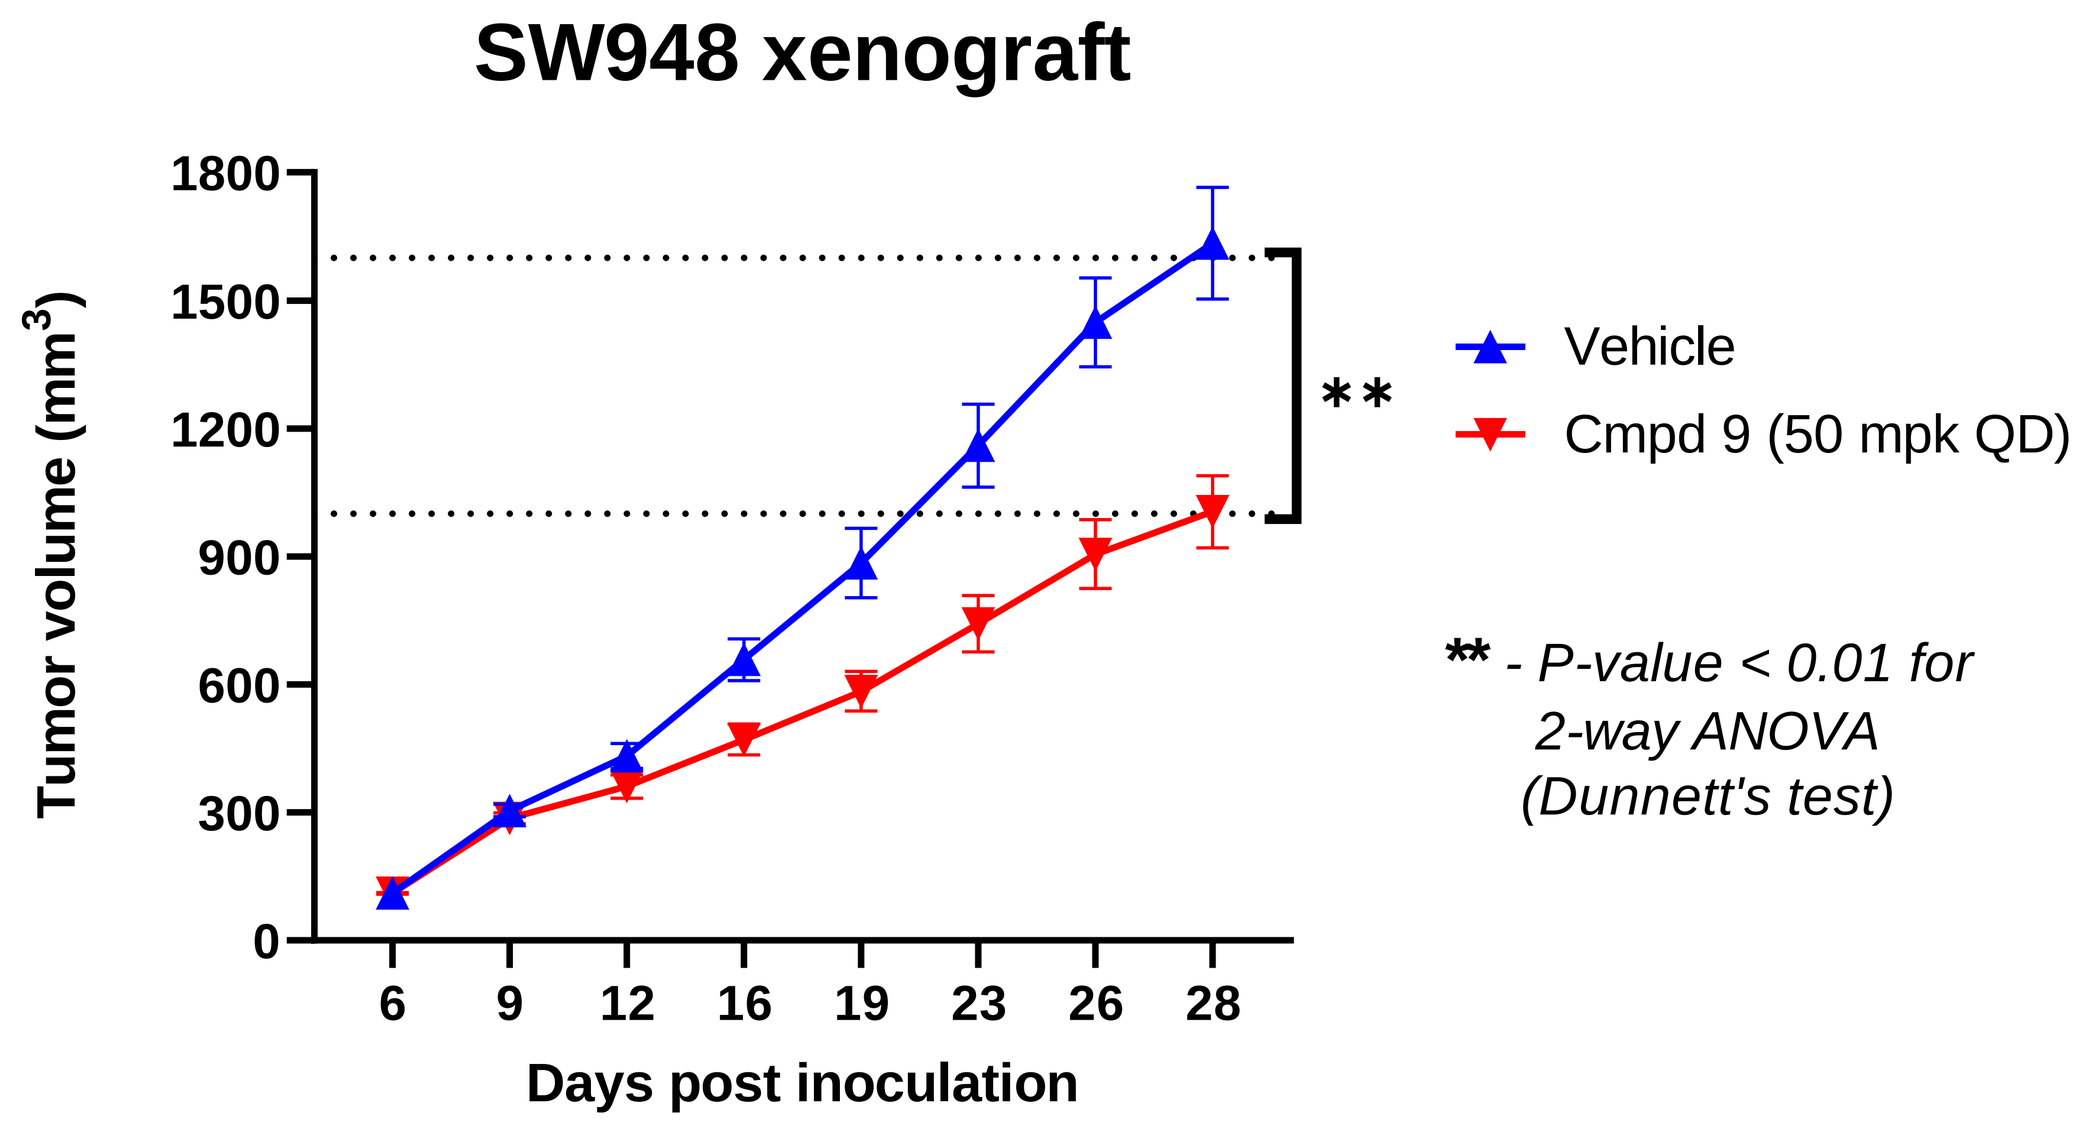
SW948 xenograft
Tumor volume (mm3)
1800
1500
1200
900
600
300
0
6
9
12
16
19
23
26
28
Days post inoculation
**
Vehicle
Cmpd 9 (50 mpk QD)
** - P-value < 0.01 for 2-way ANOVA (Dunnett's test)

Note- 150mg/Kg Loading dose administered on day 5

Conclusion:

A phenotypic screen of our library of >1000 RMAs identified hits selective for cancer cell lines over non-cancerous cell lines and demonstrated responsive SAR. The lead compound identified by this screen showed selective inhibition of total protein synthesis in colorectal cancer cell lines and induced increased expression of p21, a cell cycle regulator that is indicative of ribosomal stress. The compound selectively inhibited the growth of CRC organoids *in vitro*, and 50 mpk QD resulted in a 40% inhibition of xenograft tumor growth in mice. As a result of this data, we believe that this series of RMAs is deserving of further exploration and development as a potential anti-tumor therapy.

References:

[1]Sulima, S. O.; Hofman I. J. F.; De Keersmaecker, K.; Dinman, J. D. "How Ribosomes Translate Cancer." *Cancer Discov*. **2017**, *7*, 1069.

[2] (a)Kampen, K. R.; Sulima, S. O.; De Keersmaecker, K. "Rise of the specialized onco-ribosomes." *Oncotarget*. **2018**, *9*, 35205. (b) Genuth, N. R.; Barna, M. "The discovery of ribosome heterogeneity and its implications for gene regulation and organismal life." *Mol. Cell*, **2018**, *71*, 364.

[3]Clark, R. B.; Myers, A. G. "Discovery of Macrolide Antibiotics Effective against Multi-Drug Resistant Gram-Negative Pathogens." *Acc. Chem. Res.* **2021**, *54*, 1635.

[4]Van Rechem, C.; Black, J. C.; Boukhali, M.; Aryee, M. J.; Graslund, S.; Haas, W.; et al. "Lysine demethylase KDM4A associates with translation machinery and regulates protein synthesis." *Cancer Discov.* **2015**, *5*, 255.